H-931

Understanding and Predicting the Effect of Interleukin-7 in HIV-1 Infected Patients: A Mathematical Analysis of a Phase I/IIa Study

R. THIEBAUT[1], I. SERETI [2], J. DRYLEWICZ [1], G. TAMBUSSI [3], J. ROUTY [4], J. DELFRAISSY [5], J. MOLINA [6], M. FISCHL [7], T. CROUGHS [8], R. SEKALY [9], M. M. LEDERMAN [10], Y. LEVY [11];

1 INSERM U897, Bordeaux, France, 2 NIAID/NIH, Bethesda, MD, 3 San Raffaele Scientific Inst., Milano, Italy, 4 McGill Univ Hlth.Ctr., Montreal, Canada, 5 Hosp Kremlin Bicêtre, Paris, France, 6 Hosp St-Louis, Paris, France, 7 Univ Miami Sch. of Medecine, Miami, FL, 8 CYTHERIS, Issy Les Moulineaux, France, 9 NIML, Montreal, Canada, 10 Case Western Reserve Univ, Cleveland, OH, 11 Hosp H.Mondor, Creteil, France.

Introduction

IL-7 administration in HIV-infected patients leads to an increase of T cells (Y. Levy et al, JCI 2009, 119: 997). However, the relative contribution of production, proliferation and survival to this increase has not been quantified.

Objectives

- To estimate the contribution of the effects of IL-7 on CD4+ T cell dynamics through a mathematical analysis of observed data
- To predict CD4+ T cell response to IL-7 administration

Methods

Study design

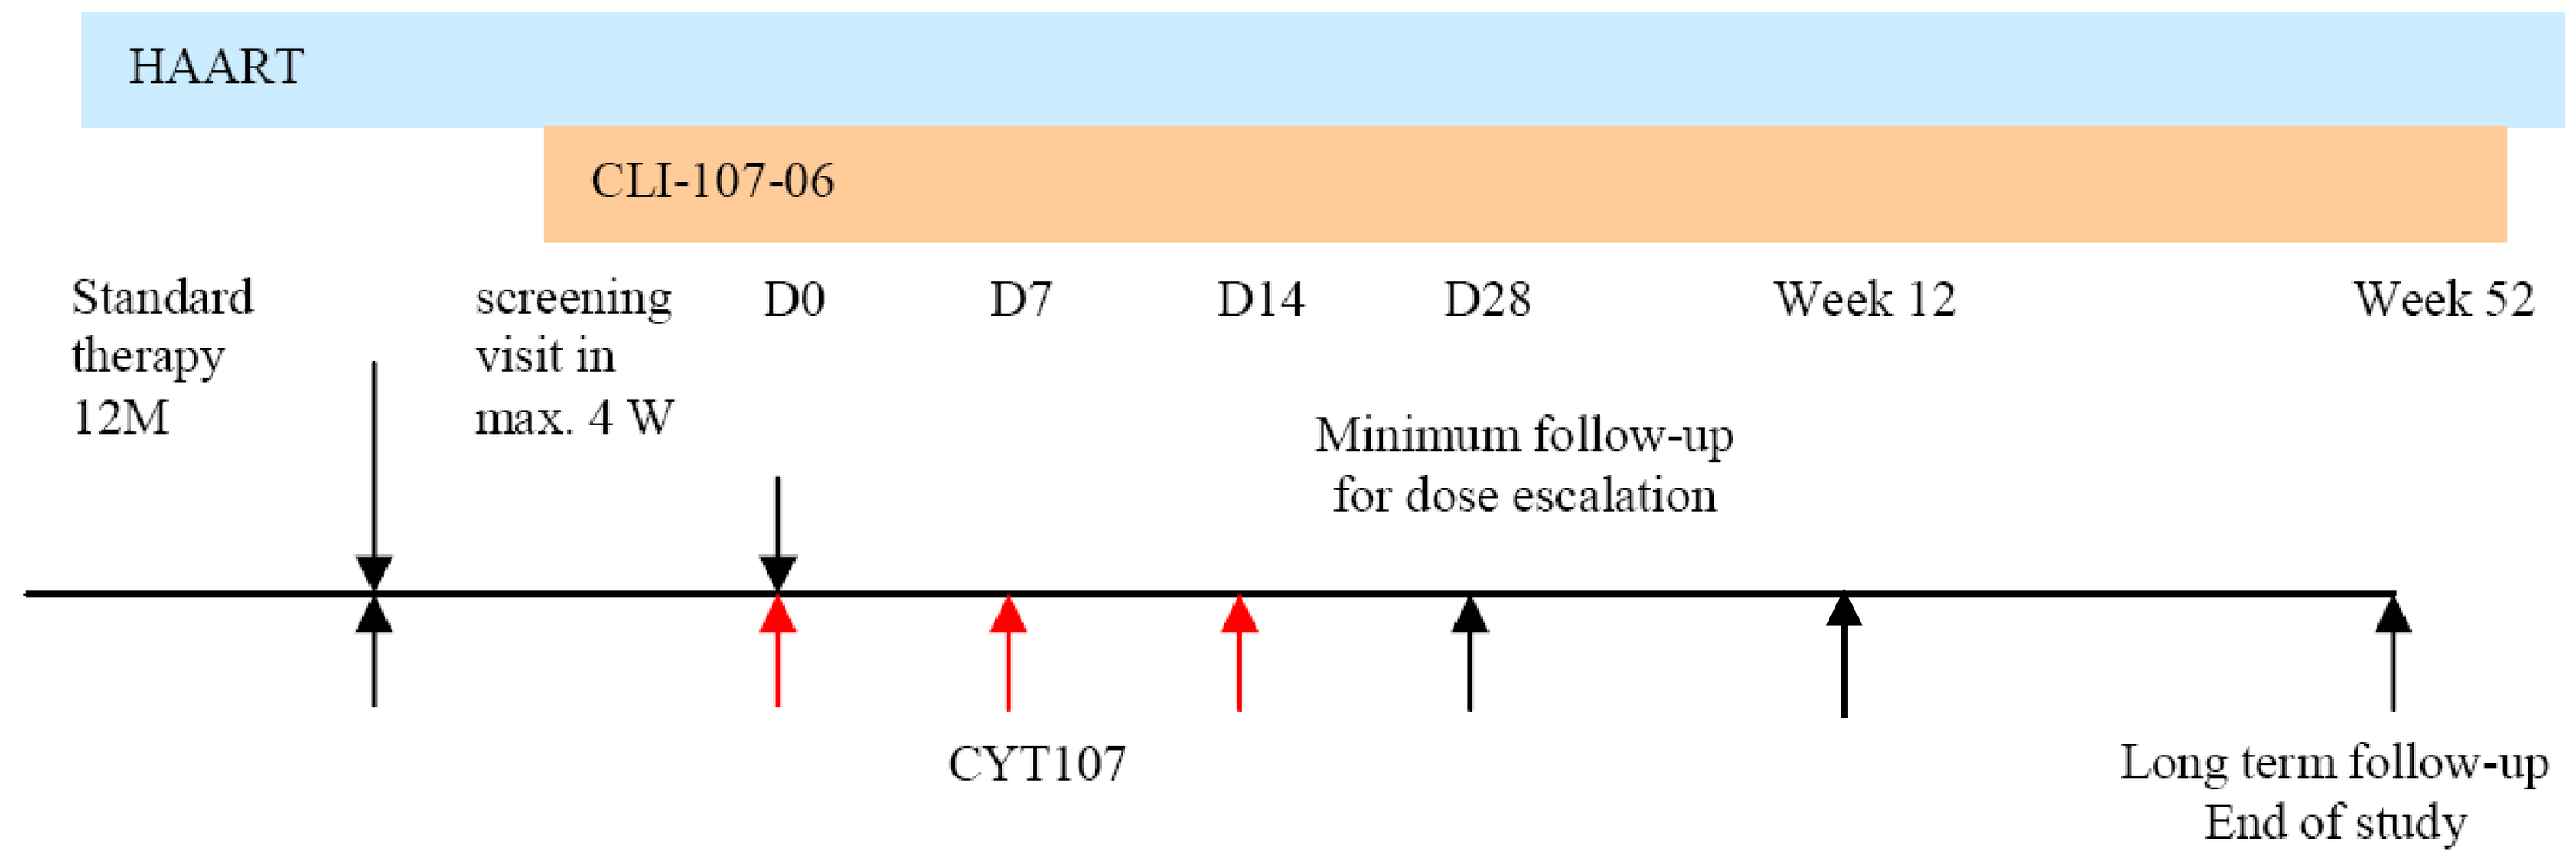
HAART
CLI-107-06
Standard therapy 12M
screening visit in max. 4 W
D0
D7
D14
D28
Week 12
Week 52
Minimum follow-up for dose escalation
CYT107
Long term follow-up End of study

(red arrows: CYT107 injection, black arrows: sampling times)

Data came from a dose-escalation study of glycosylated IL-7 (CYT107, 1 dose/week for 3 weeks) in HIV-infected patients with CD4+ T cell count between 101 and 400 cells/µL and plasma HIV RNA< 50 cp/mL while on ART (Levy et al. ICAAC 2009, abs H-1230a). Eight patients were randomized to CYT107 and two to placebo at each dose level (10, 20 and 30 µg/kg/week) and were followed until week 52.

Contact: R Thiébaut, INSERM U897, Bordeaux, France
Rodolphe.thiebaut@isped.u-bordeaux2.fr

Description of the model

We developed a 2 compartment mathematical model comprising the quiescent (Ki67-) and the proliferating (Ki67+) CD4+ T cells. The reversion rate (ρ) could be approximated by the inverse of the duration of KI67 expression. A maximum likelihood approach was used to fit this model to the observed data.

Model for CD4+Ki67- (non proliferating) and CD4+Ki67+ (proliferating) T cells

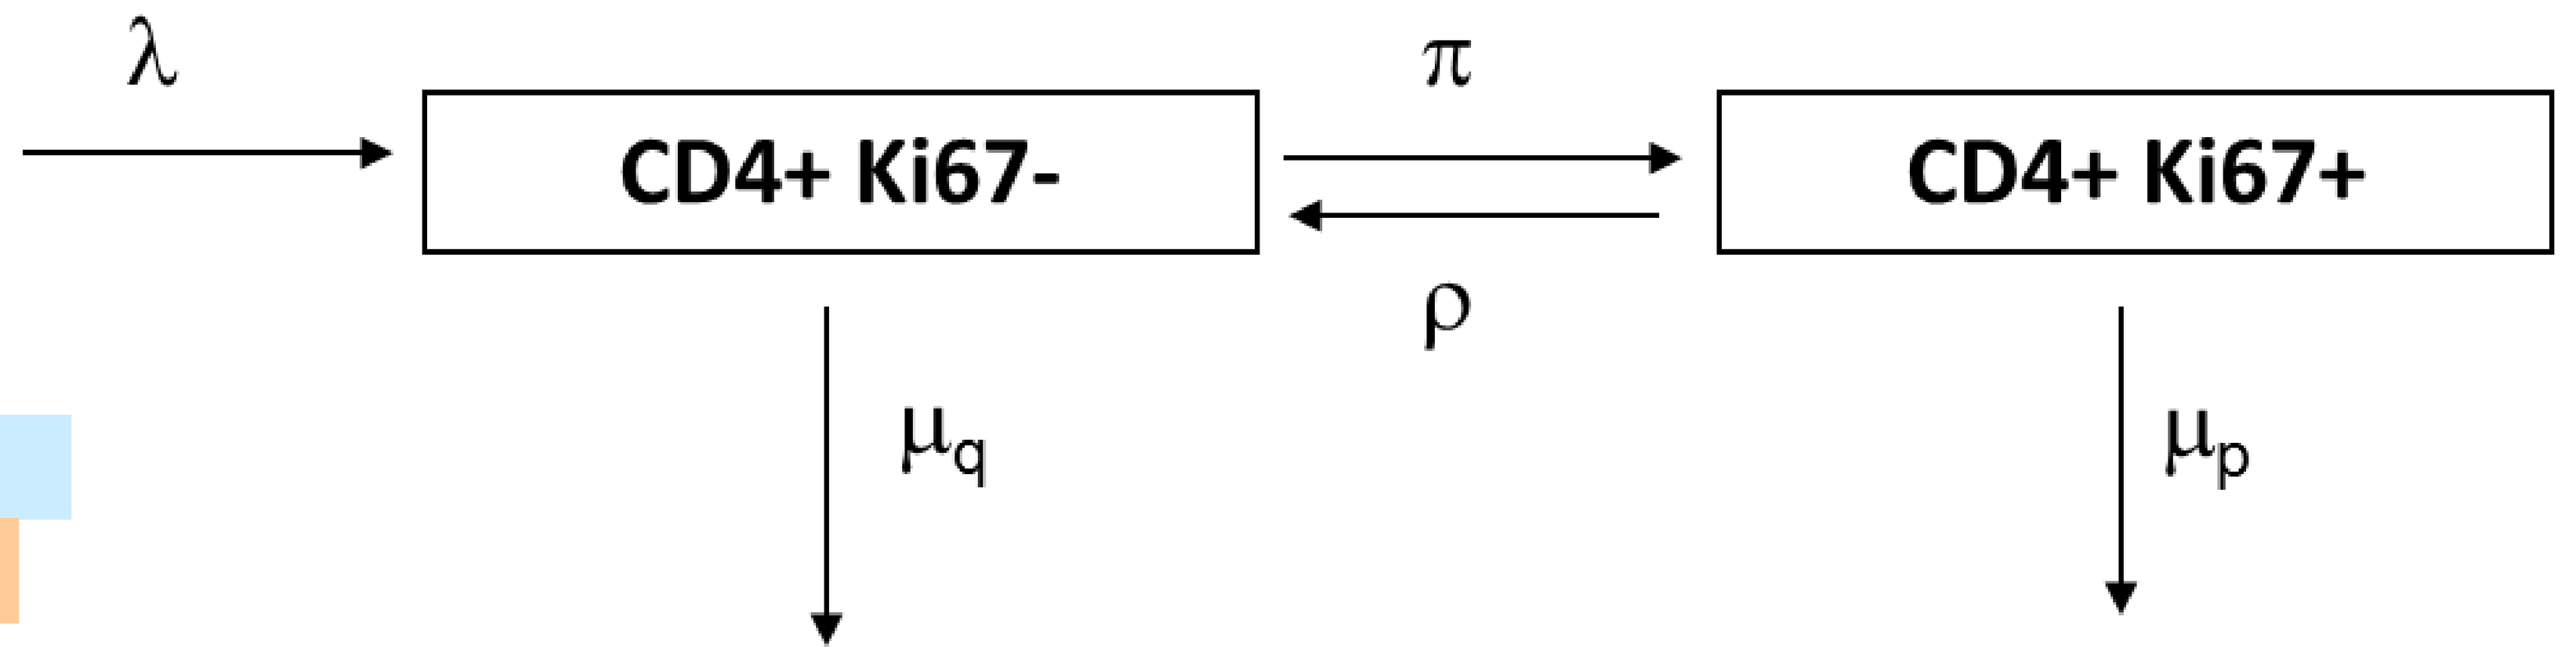
λ
CD4+ Ki67-
π
ρ
CD4+ Ki67+
μq
μp

IL-7 effect was evaluated on the production rate of Ki67- (λ), the proliferation rate (π) and the loss rates of Ki-67- cells (μq). Hence each parameter was allowed to change over time (i.e. before, during and after IL-7 administration). Several models have been compared and the best models are presented (according to the AIC statistical criteria: the best model having the lowest AIC value). The effect of IL-7 could not be checked on more than two parameters at the same time with the available data.

Two models are presented:

- Model 1: Effect of IL-7 on proliferation rate only (parameter π)
- Model 2: Effect of IL-7 on proliferation and loss rates (parameters π and μq)

Results

A dose-dependent increase of CD4 T cell counts was observed in IL-7 treated participants

(observed median CD4+ T cells/µL by group)

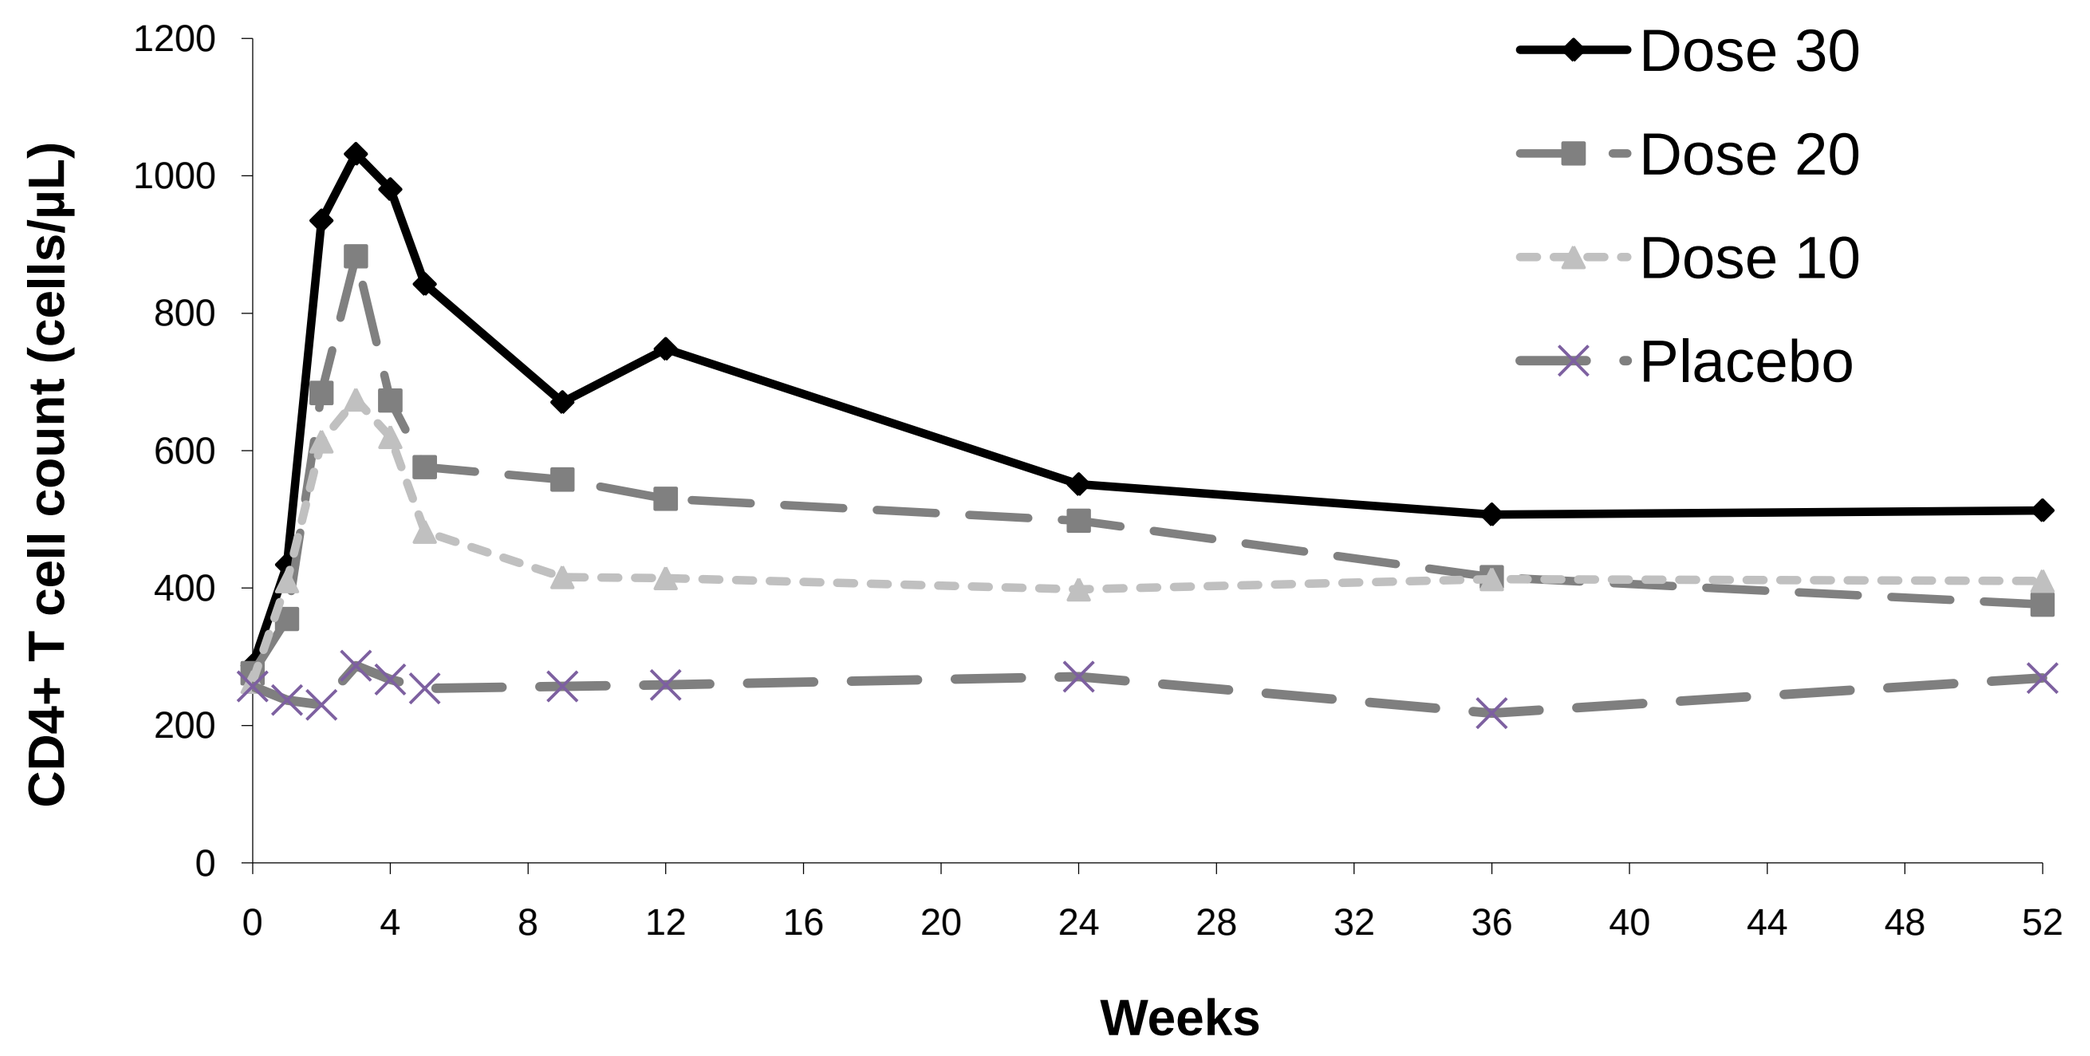
CD4+ T cell count (cells/µL)
Weeks
Dose 30
Dose 20
Dose 10
Placebo

IL-7 led to a dose-dependent increase of the proportion of CD4+ T cell expressing Ki67

(observed median fold increase of %Ki67+ by group)

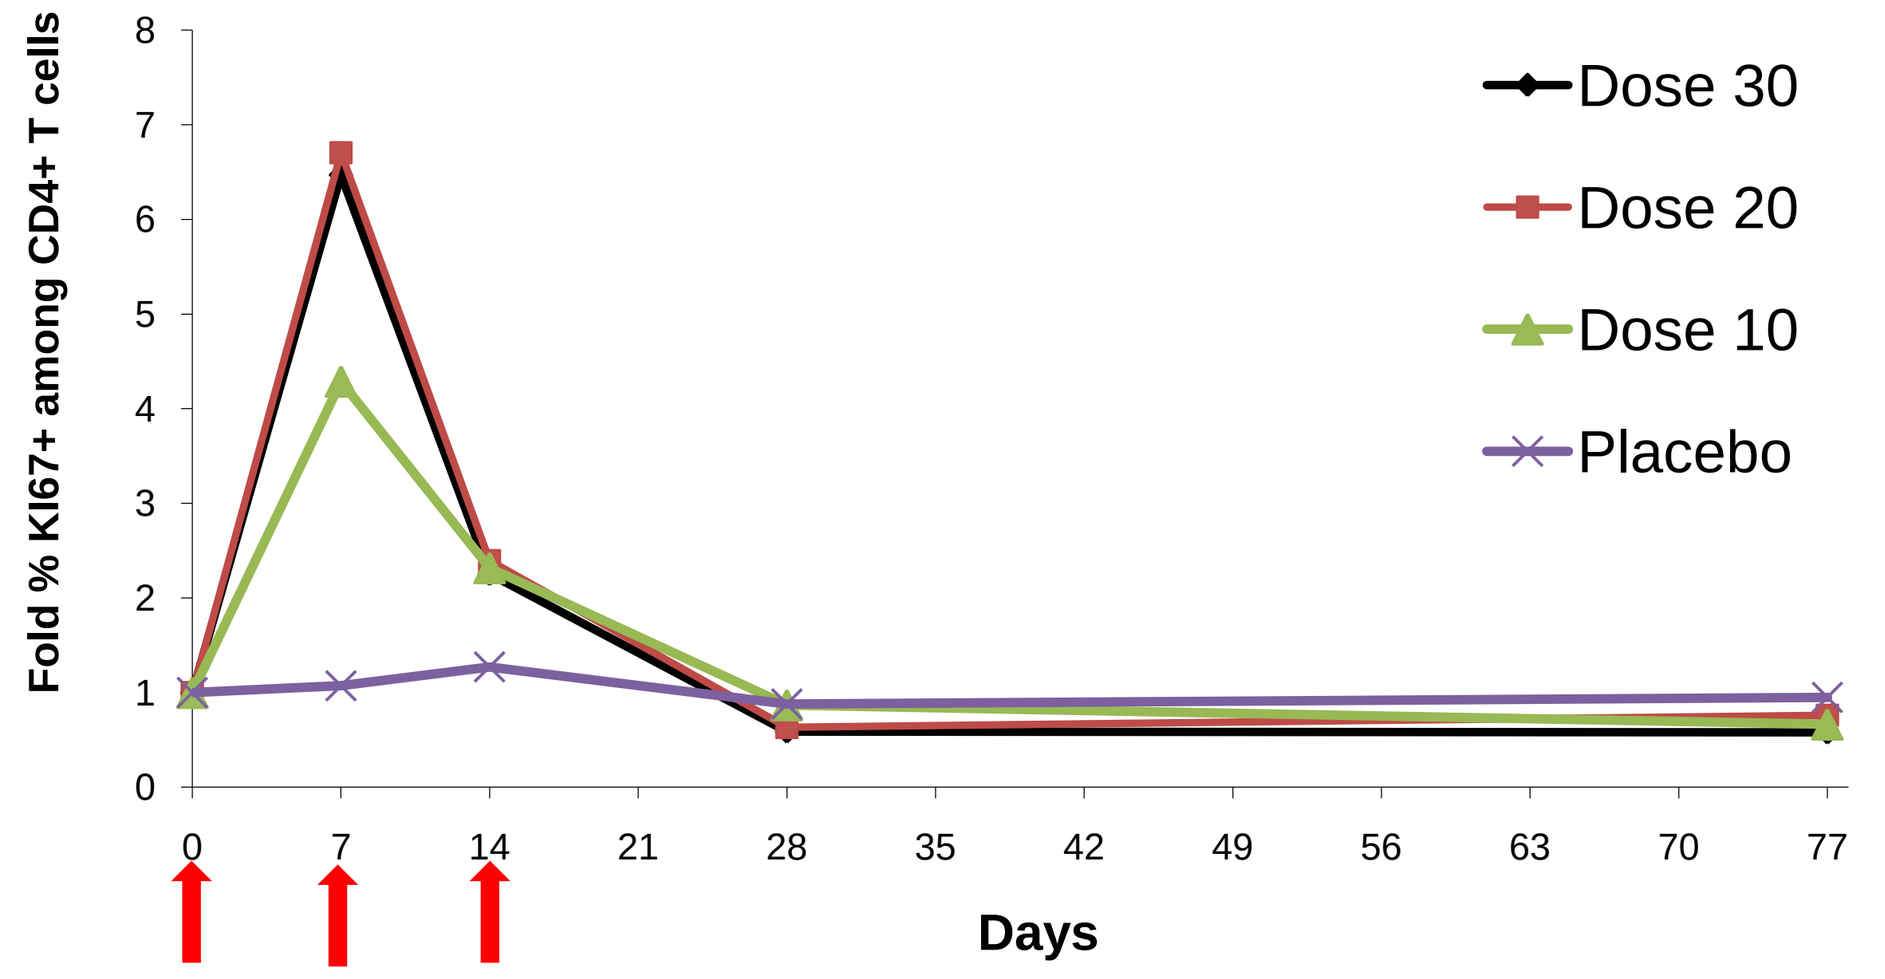
Fold % KI67+ among CD4+ T cells
Days
Dose 30
Dose 20
Dose 10
Placebo

Examples of individual fits from model 2

(triangle: observed data, plain line: model prediction)

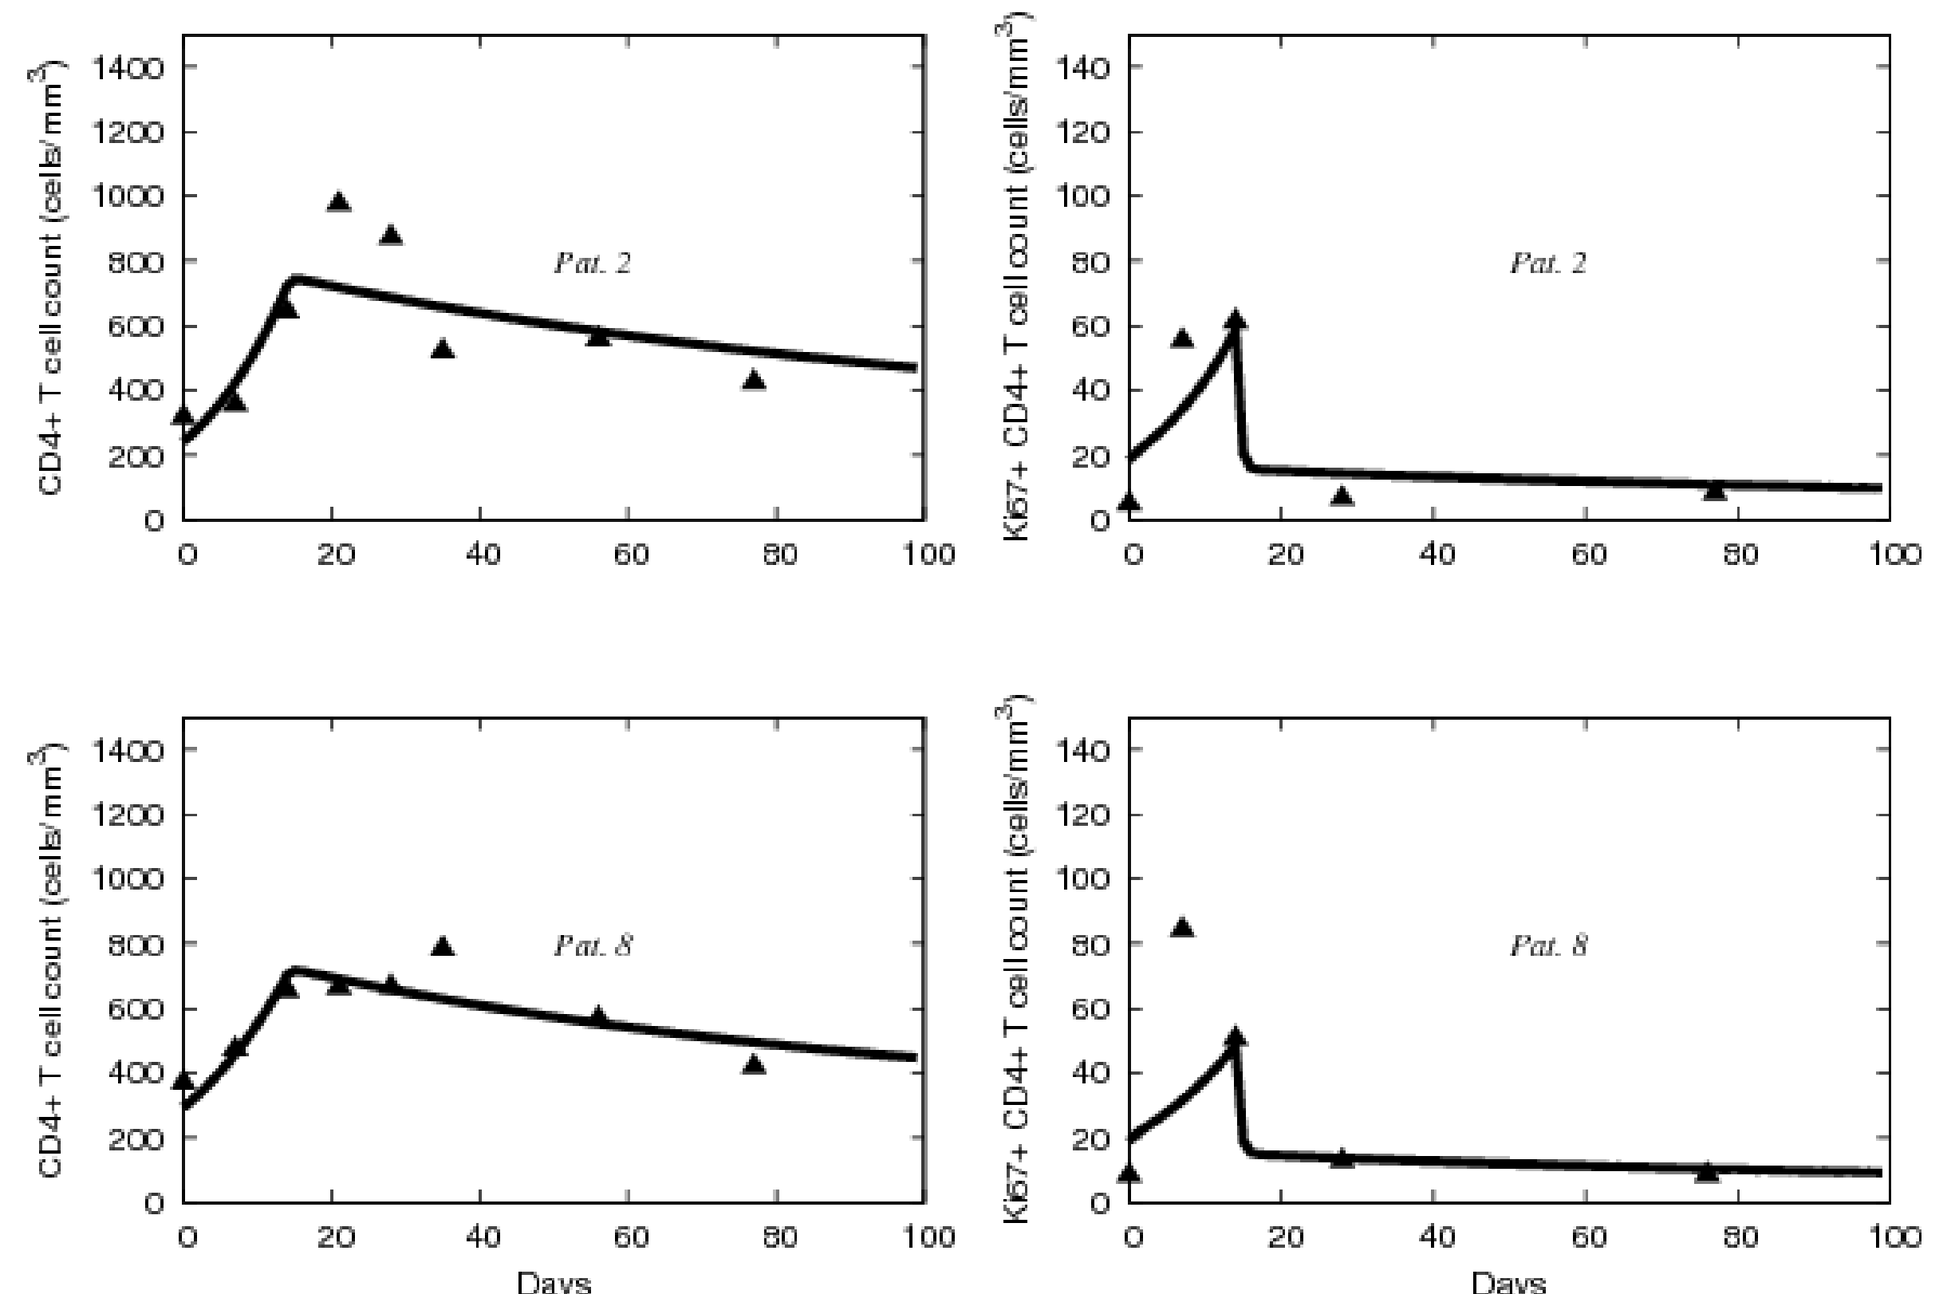
CD4+ T cell count (cells/mm³)
Ki67+ CD4+ T cell count (cells/mm³)
Pat. 2
Pat. 8
Days

Estimates of model parameters

	Parameters	Model 1 (AIC 168)	Model 2 (AIC 136)
l	Constant production rate	3.18	12.0
p	Proliferation rate		
	- Before & after CYT107	0.024	0.021
	- During CYT107: * dose 10	0.085	0.123
	* dose 20	0.122	0.140
	* dose 30	0.160	0.158
μ_q	Loss rate of non proliferating cells		
	- Before & during CYT107	0.030	0.063
	- After CYT107	0.030	0.047
μ_p	Loss rate of proliferating cells	0.163	0.215

The best fit was achieved when the IL-7 effect was considered on both proliferation and loss rates (AIC 136 vs. 168 for the effect on proliferation only). The proliferation rate increased during IL-7 administration with a linear dose effect (.02 day-1 at baseline, .12, .14, .16 with 10, 20 and 30 µg/kg/week, $p<.001$). The loss rate of CD4+Ki67- T cells decreased with IL-7 treatment but the decrease was not dose-dependent (from .063 d-1 to .047, $p<.01$).

Conclusion

- This analysis of CD4 T cell dynamics under IL-7 therapy suggests that IL-7 exerts a transient and strong effect on peripheral T cell proliferation and also a significant improvement in cell survival as found in another phase I study (Thiébaut et al., CROI 2010, abs 389).
- Future studies are designed to provide additional information (e.g. naïve cells, recent thymic emigrant) in order to evaluate the effects of IL-7 on T cell production rates.